Diagnosis and treatment of individuals with MPS II Hunter disease in the United Kingdom[(B)]

Sophie Thomas,[1] Alex Morrison.[1]

[1]The Society for Mucopolysaccharide Diseases (MPS Society), Buckinghamshire, UK

Introduction

- Hunter disease (MPS II) is a rare, X-linked disease which is caused by a deficiency of iduronate-2-sulphatase, leading to a progressive accumulation of glycosaminoglycans in cells, tissues and organs.[1,2]
- Individuals with MPS II experience a range of clinical manifestations, including: airway obstruction, skeletal deformities and cardiac disease. Individuals with central nervous system (CNS) involvement, experience cognitive impairment and neurological decline.[1,2]
- MPS II signs and symptoms typically manifest between 18 months and 4 years in individuals with the severe phenotype; this is further delayed by around 2 years in the attenuated phenotype.[3]
- The first diagnostic approach, and the one that patients and their carer's generally remember, is urinary GAG assessment.
- Urinary analysis can indicate the presence of an MPS disorder, but a definitive MPS II diagnosis is via blood enzyme analysis; the best practice followed in the UK is that a positive urinary GAG result is followed by a blood test or genetic testing.
- Prenatal diagnosis is available for foetuses at risk of MPS II.[1]
- Whilst enzyme replacement therapy (ERT) has been shown to improve the signs, symptoms and wellbeing of individuals with MPS II;[1] data on haematopoietic stem cell transplantation (HSCT) are rare.[4]
- The aim of this project was to understand how and when MPS II is diagnosed and treated in the UK and to determine the prevalence of CNS involvement and concomitant diagnoses.

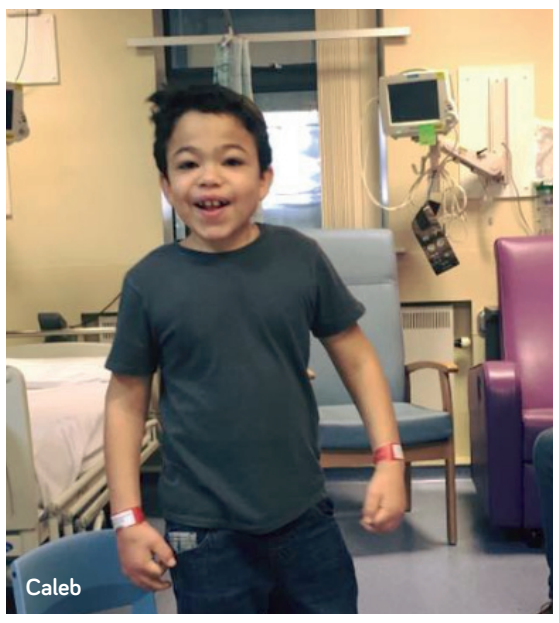
Caleb

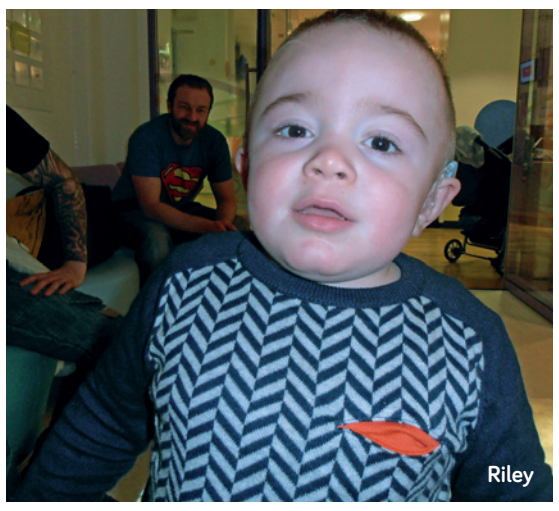
Riley

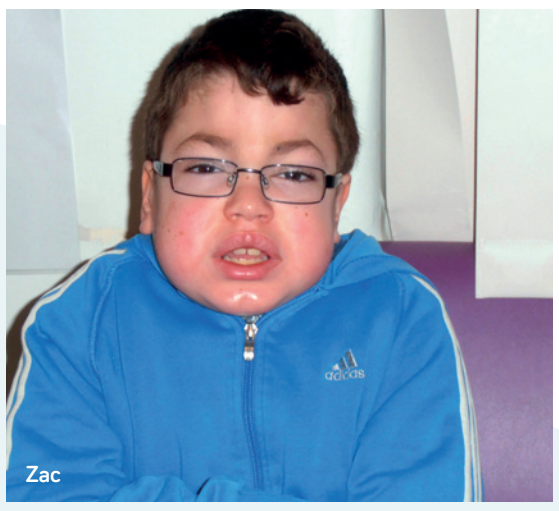
Zac

Methods

- Seventy-one individuals with MPS II resident in the UK were identified by the MPS Society and invited to take part in the survey via telephone interview.
- A specifically designed questionnaire was used to assess the individual's diagnoses and treatment as well as their educational attainment and need for support from primary through to further education.
- Interviews took place in December 2015 and January 2016.
- Results for the diagnosis and treatment section of the survey are presented here.
- Results for the educational attainment and support from primary through to further education are presented in Poster 164.

Results

Diagnosis

- Forty-one individuals agreed to take part in the study (58%), ranging in age from 1 to 36 years (mean 12.3 years).
- Age at diagnosis of MPS II ranged from 6 days to 7 years (mean 2.5 years).
- Diagnosis was earlier in the younger individuals (under 8 years), with a mean of 1.8 years (range 6 days to 3 years), compared to a mean of 2.9 years (range 6 months to 7 years) in those aged 8 years and over.
- Sixty-five percent of individuals had seen other physicians or specialists before their MPS II diagnosis (Figure 1).

Figure 1. Physician and specialist involvement before MPS II diagnosis

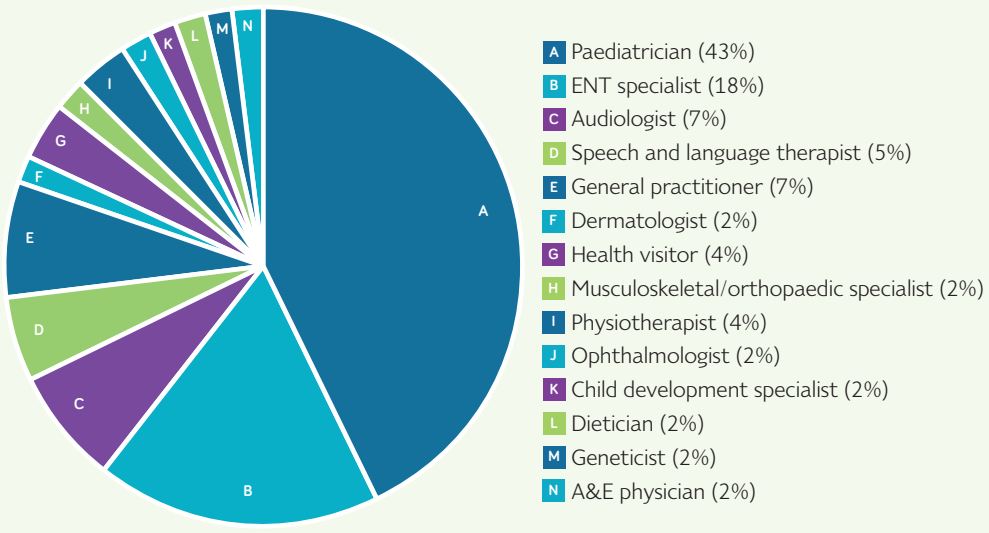
A Paediatrician (43%)
B ENT specialist (18%)
C Audiologist (7%)
D Speech and language therapist (5%)
E General practitioner (7%)
F Dermatologist (2%)
G Health visitor (4%)
H Musculoskeletal/orthopaedic specialist (2%)
I Physiotherapist (4%)
J Ophthalmologist (2%)
K Child development specialist (2%)
L Dietician (2%)
M Geneticist (2%)
N A&E physician (2%)

- Thirty-one percent of individuals questioned had seen more than one specialist before receiving a diagnosis of MPS II.
- In our cohort there were no reports of pre-natal diagnosis (chorionic villus sampling or amniocentesis).
- The majority of diagnoses (61%) were made via a combination of blood and urine enzyme analysis (Figure 2).

Figure 2. Method of MPS II diagnosis

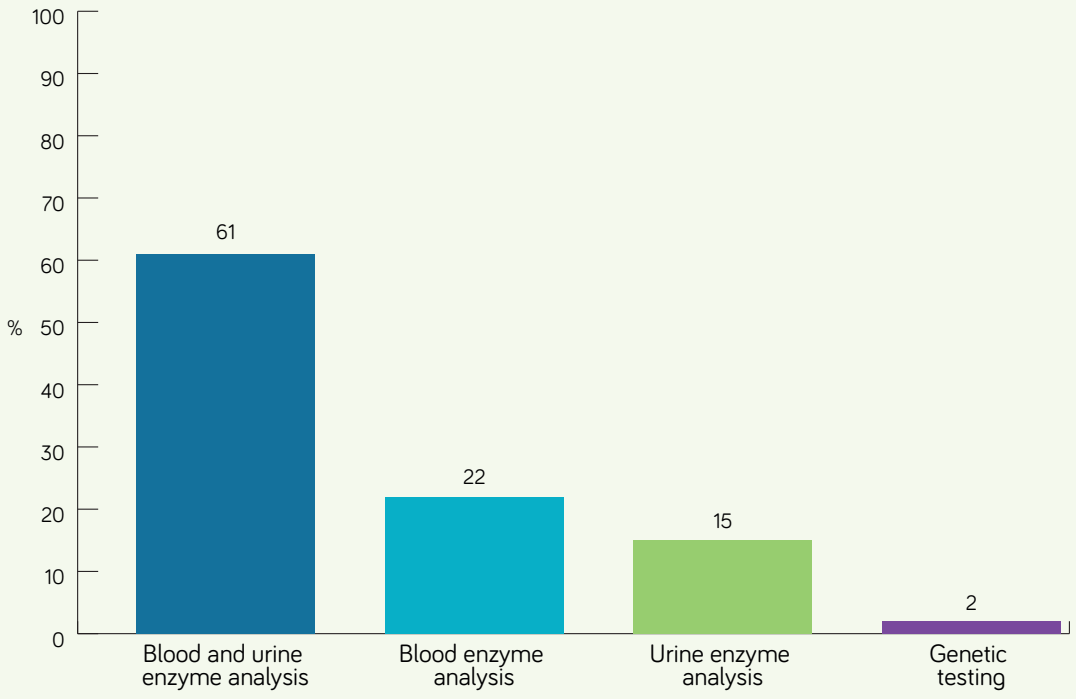
%
100
90
80
70
60
50
40
30
20
10
0
61
22
15
2
Blood and urine enzyme analysis
Blood enzyme analysis
Urine enzyme analysis
Genetic testing

- There was one report genetic testing (2%) which had taken place outside of the UK. This took place in the absence of blood and/or urine enzyme analysis.
- One individual reported hair/stool analysis as part of their MPS II diagnosis; this was carried out in addition to blood and urine enzyme analysis.
- Concomitant diagnoses were made in 7 individuals (17%). Attention deficit hyperactivity disorder and autistic spectrum disorder were the most prevalent concomitant diagnoses (Figure 3); all concomitant diagnoses were made after the diagnosis of MPS II.

Figure 3. Concomitant diagnoses following MPS II diagnosis

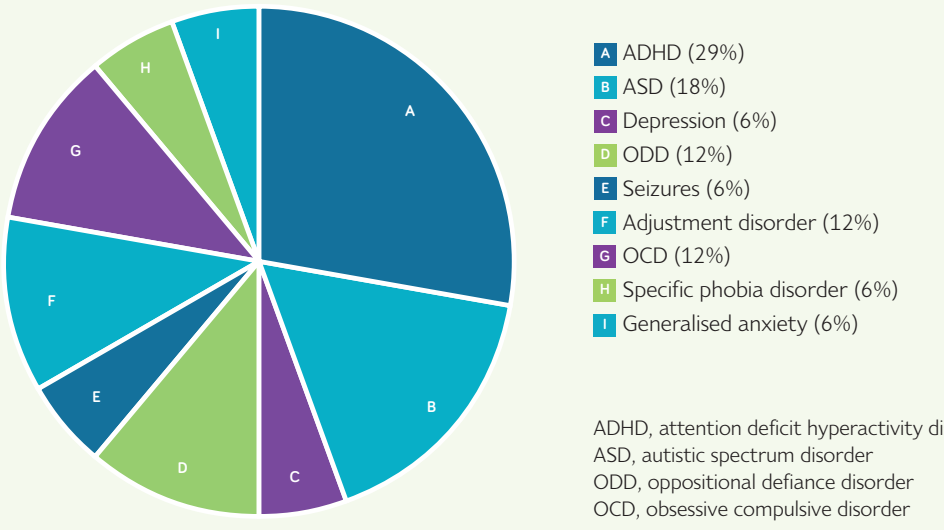
A ADHD (29%)
B ASD (18%)
C Depression (6%)
D ODD (12%)
E Seizures (6%)
F Adjustment disorder (12%)
G OCD (12%)
H Specific phobia disorder (6%)
I Generalised anxiety (6%)

ADHD, attention deficit hyperactivity disorder
ASD, autistic spectrum disorder
ODD, oppositional defiance disorder
OCD, obsessive compulsive disorder

- Of the 41 individuals surveyed, 54% (n=22) reported CNS involvement; 37% (n=15) reported no CNS involvement; four individuals (10%) did not know whether there was CNS involvement or not.
- A review of all responders data indicated all but 3 had some level of CNS involvement; 49% (n=20) had severe progressive CNS involvement.

Treatment

- Ninety percent (n=37) of individuals were being or had been treated with ERT, including individuals receiving or who had received intravenous ERT and those participating in a clinical trial for intrathecal ERT.
- Overall, the mean age at start of ERT was 5.9 years (range 8 weeks to 27 years).
- Children born since the reimbursement of ERT (2007 in England, Scotland and Northern Ireland) started ERT earlier than the overall population, with a mean age of 2.4 years (range 8 weeks to 4 years).
- Four individuals (10%) received a HSCT; 3 individuals were under 1 year (8 weeks, 4 months and 10 months); the other aged 2 years and 3 months, at the time of the transplant.
- Half of the individuals who received a HSCT, also received ERT at some point in their treatment.
- HSCT consisted of bone marrow (n=2) and cord blood (n=2). No individuals received mobilised peripheral blood stem cells.
- One individual (2%) had not received any form of treatment for MPS II.

Conclusions

- **MPS II is being diagnosed at an earlier age, but in the UK it can still take up to 3 years to obtain a definitive diagnosis. This is in line with other published findings.[3]**
- **Individuals are often referred to one or more specialists before an MPS II diagnosis is reached; with specialist involvement dependent upon presenting features.**
- **Most individuals are being treated with ERT and following the reimbursement of ERT in 2007, normally start treatment before the age of 3.**
- **In agreement with the literature, only a small proportion of individuals in the UK receive a transplant as part of their treatment for MPS II.[4]**

The data supporting this poster were gathered via a questionnaire filled in by patients or their carer's.

The accuracy of the data in these patient reported outcomes cannot be confirmed without consulting the respective patient medical records.

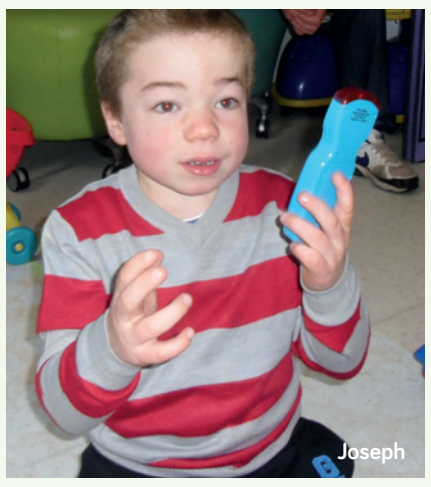
Joseph

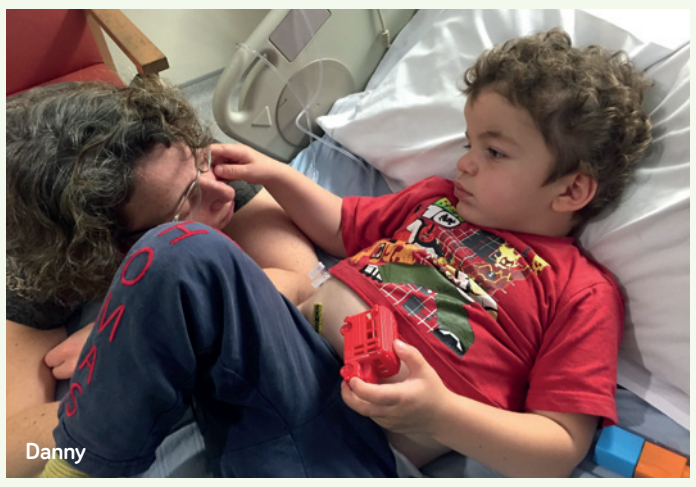
Danny

References

1. Wraith J, et al. 2008. Eur J Pediatr;167:267.
2. Muenzer J, et al. 2012. Eur J Pediatr;171:181.
3. Muenzer J, et al. 2009. Pediatrics; 124:e1228.
4. Giugliani R, et al. 2014. Genet Mol Biol;37:315.

Acknowledgements

- Rebeca Brandon, MPS II Advocacy Officer at the MPS Society, for all of her support and work with individuals and their families.
- Jaromir Miki (Shire) and Meng Wang (Evidera) who were active participants in the design of the questionnaire.
- Patient questionnaires were conducted by Jo Goodman and Benedicta Marshall-Andrew (MPS Commercial) and Alex Cavell.
- Medical writing and editorial support was provided by Jacqueline Adam, PhD, MPS Commercial.
- This research was funded by Shire Human Genetic Therapies, Inc.

MPS support research awareness Wicked GENES

Society for Mucopolysaccharide Diseases
www.mpssociety.org.uk